PCV38

ISPOR 19th Annual European Congress

Vienna, October 29th | November 2nd 2016

BUDGET IMPACT ANALYSIS OF COSYREL FOR THE TREATMENT OF HYPERTENSION IN ITALY

Orlando VL[1], Lanati E[1], Doratori S. [2]

[1]MA Provider, Milan, Italy

[2]Servier Group, Rome, Italy

Introduction: **Cardiovascular diseases (CVDs)** are the leading causes of death and disability in the world, both in low and middle-income countries and in high-income countries[1].
Hypertension is a major risk factor for ischemic and hemorrhagic stroke, myocardial infarction (MI), heart failure (HF), chronic kidney disease, cognitive decline and premature death. In 2008, approximately 40% of adults aged ≥25 years had been diagnosed with hypertension worldwide[2].
Coronary artery disease (CAD) is the most common type of heart disease, a leading cause of death, morbidity and loss of quality of life globally, predicted to remain so for the next 20 years, making it a major public health problem. CAD is the underlying cause of heart failure in about 65% of patients. Each year, approximately 3.8 million men and 3.4 million women die from CAD[3].
Heart failure is a major cause of morbidity and mortality in Europe and the United States. Over 26 million people suffer from heart failure around the world and over 3.5 million people are newly diagnosed with heart failure every year in Europe alone[4].
According to the Italian Hypertension Society (SIIA), in Italy there are approximately 14 million of people who have elevated blood pressure (hypertension), but only 3 million receive appropriate therapy and just the 37% of these with controlled blood pressure.

		Prescriptions	Patients
Dual therapy	Perindopril + Beta blockers	667.548	55.629

Ex factory prices	Price per day	Price per month
Bisoprolol	€ 0,09	€ 2,70
Perindopril	€ 0,13	€ 3,90
Perindopril + Bisoprolol	€ 0,22	€ 6,60
Nebivolol	€ 0,18	€ 5,49
Carvedilol	€ 0,15	€ 4,36
Perindopril + Nebivolol	€ 0,31	€ 9,39
Perindopril + Carvedilol	€ 0,28	€ 8,26
Average price of dual therapy with Perindopril + Beta blockers	€ 0,27	€ 8,08
Cosyrel	€ 0,21	€ 6,27

Table 1. **Upper panel**: Patients in double therapy (IMS)
Lower panel: Ex-factory prices (AIFA)

Objectives: The objective of this study was to perform a Budget Impact Analysis (BIA) assessing the introduction of Cosyrel for the treatment of hypertension into the Italian market. COSYREL is a single pill combination of a beta-blocker (bisoprolol fumarate) and an angiotensin-converting enzyme inhibitor (perindopril arginine) indicated as substitution therapy for treatment of hypertension and/or stable coronary artery disease and/or stable chronic heart failure with reduced systolic left ventricular function in adult patients adequately controlled with bisoprolol and perindopril.

Methods: The BIA compares two different scenarios: one without a dual fixed combination therapy of Perindopril and Bisoprolol (Scenario 1) vs. another one with the introduction of Cosyrel (Scenario 2). The study has been conducted considering the perspective of the Italian National Healthcare Service. Population data were obtained from IMS database (Medical Audit Q4 2015 MAT 12 months). The time horizon considered was 3 years from the introduction of Cosyrel. Total number of patients in each of the 3 years was the same for the two Scenarios, because the model allows only the switch of patients from the dual combinations (Perindopril+Beta blockers) to the fixed dose treatment with Cosyrel.
According to the IMS database, there are 667.548 prescriptions of dual combination of Perindopril and Beta blockers, equal to about 56 thousand patients treated (Table 1). The reference market for Cosyrel is composed by all the patients treated with the double therapy, considering a yearly incidence rate equal to 2% (IMS)

The model assumes that the percentage of patients who switch to Cosyrel is 55%, 70% and 80% the first, second and third year respectively.
Prices used in the model are ex-factory prices (Table 1) as published in AIFA web site after price cuts.
The total cost of the dual therapy (perindopril + beta blockers) in the first scenario has been calculated using the average price of the dual therapies of perindopril associated with bisoprolol, nebivolol or carvedilol, representing the 80% of the Italian beta blockers market.

Results: The study shows that the introduction of Cosyrel leads to a reduction in the quantity of package (1.365832, 1.397.264 and 1.429.419 in Scenario 1 and 990.228, 908.222 and 857.651 in Scenario 2, respectively in year 1, 2 and 3). (Figure 1) With a cost of 6,27 €/month, the introduction of Cosyrel generates savings for the Italian NHS equal to 680.594, 886.145 and 1.036.043 € respectively in year 1, 2 and 3 over the total expenditure of 5.519.328, 5.646.344 and 5.776.283 € related to the year 1, 2 and 3 in the first scenario (Figure 2). As the majority of patients with hypertension require two or more agents to achieve their BP targets, simplifying treatment by reducing pill burden through the use of SPCs is one of the most straightforward and effective ways of improving adherence[5].

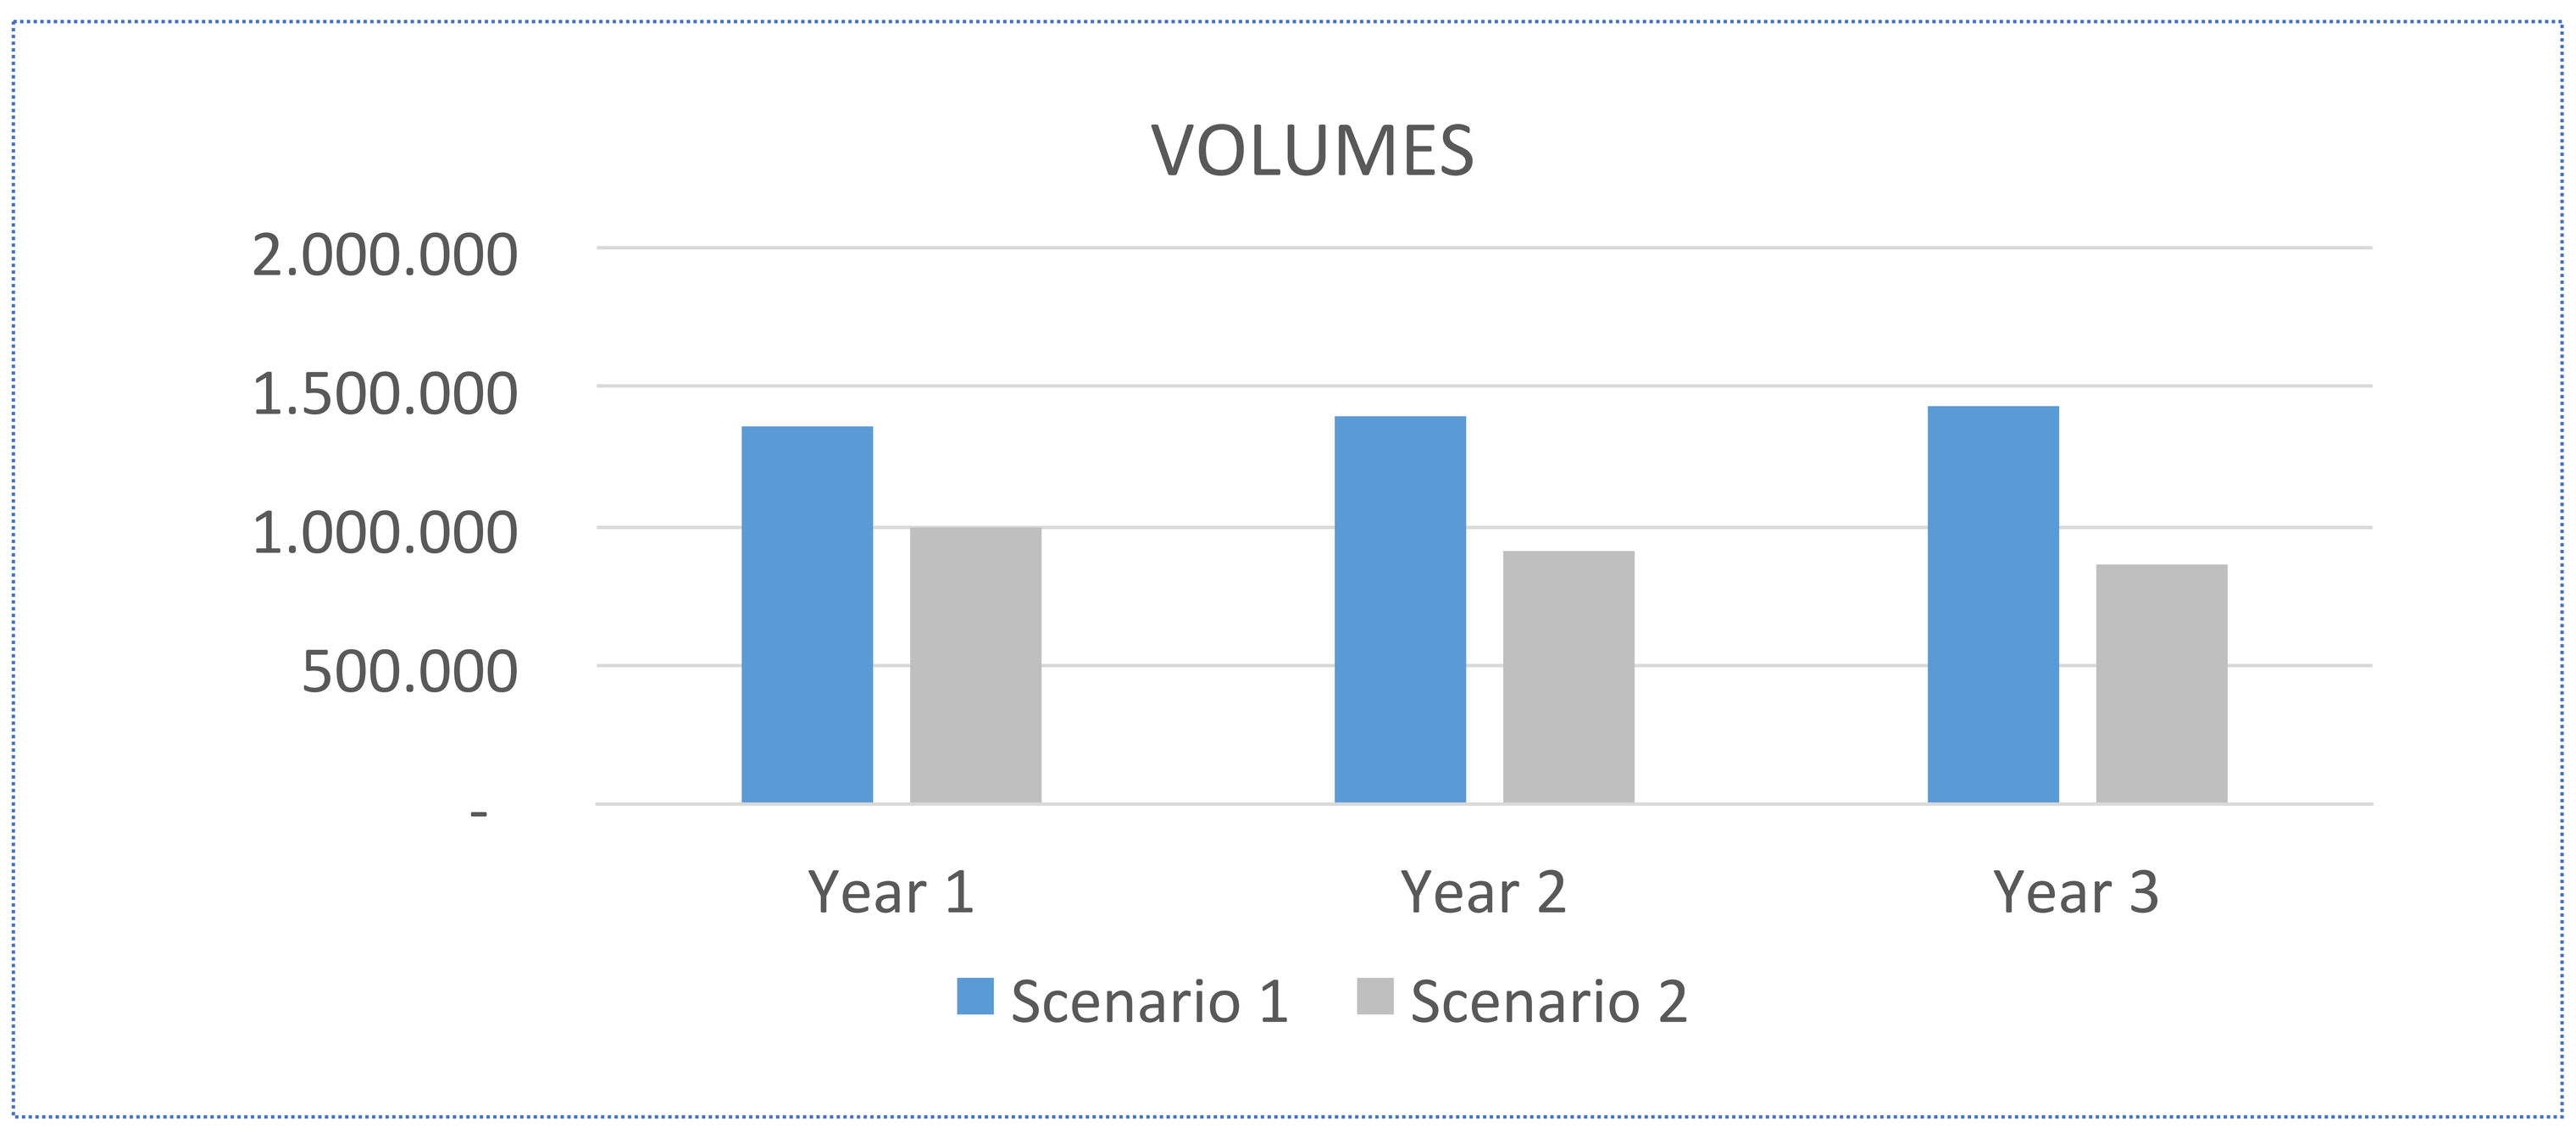
VOLUMES
2.000.000
1.500.000
1.000.000
500.000
-
Year 1
Year 2
Year 3
Scenario 1
Scenario 2

Figure 1

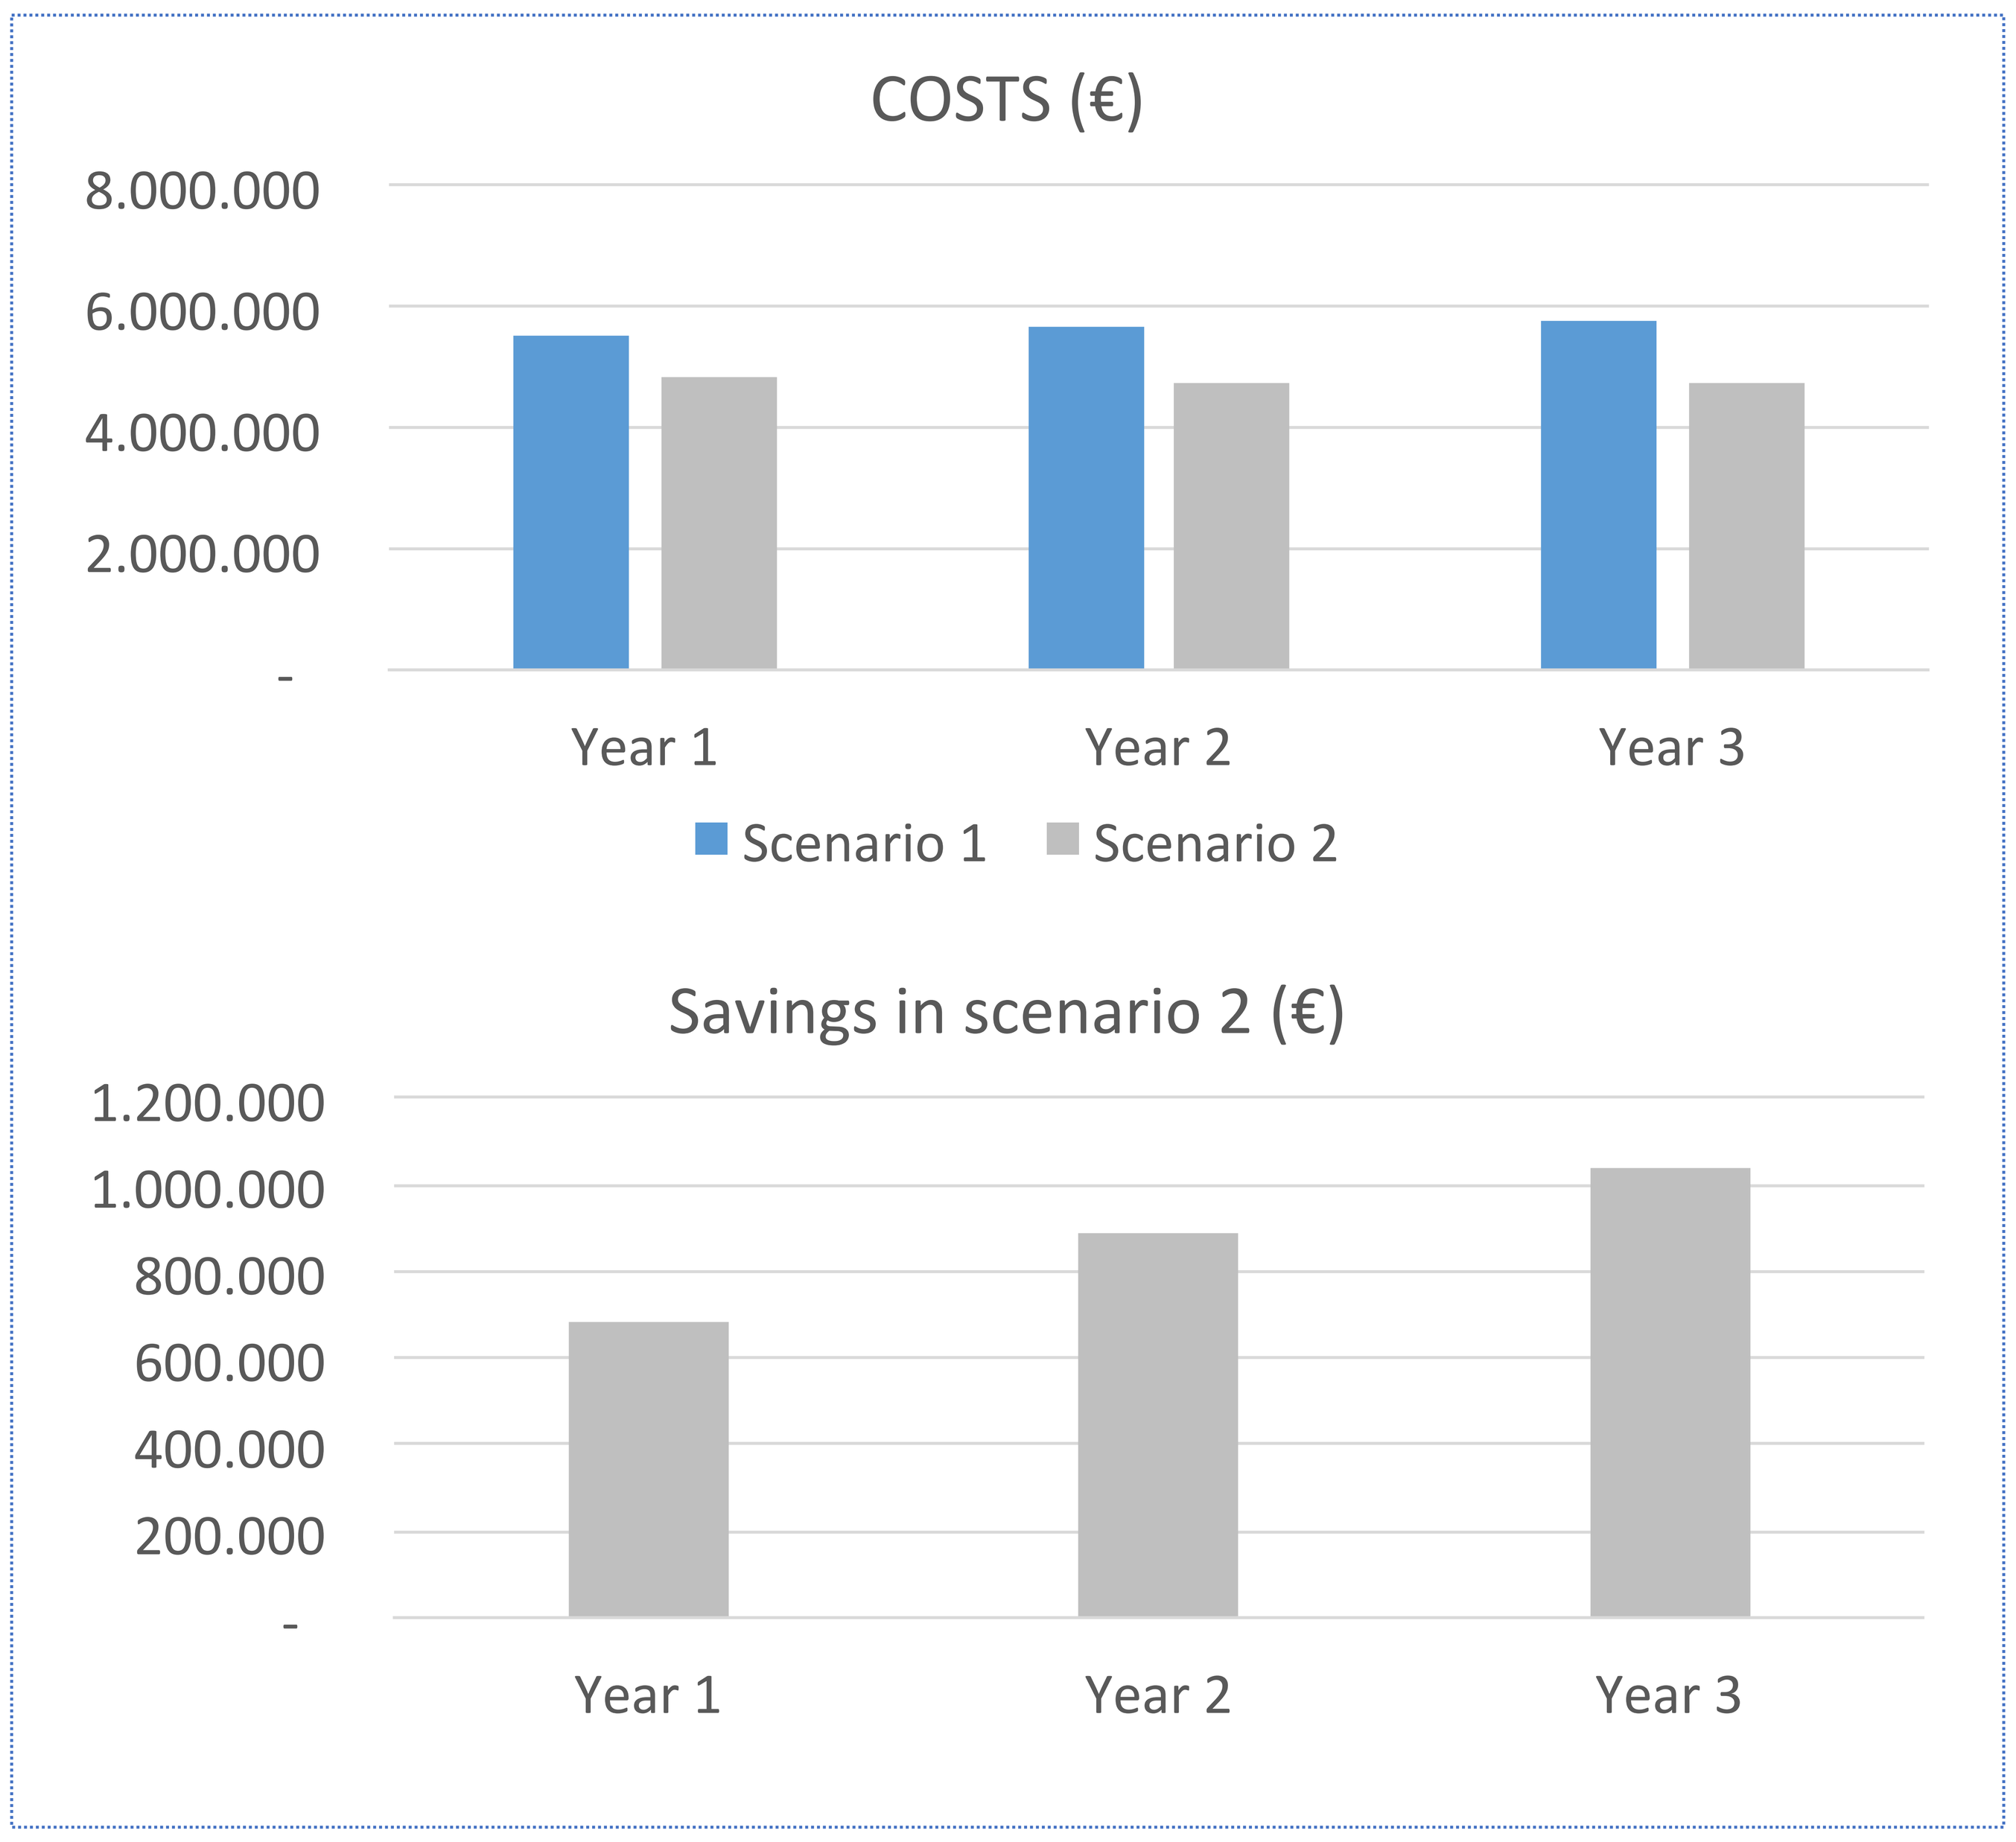
COSTS (€)
8.000.000
6.000.000
4.000.000
2.000.000
-
Year 1
Year 2
Year 3
Scenario 1
Scenario 2
Savings in scenario 2 (€)
1.200.000
1.000.000
800.000
600.000
400.000
200.000
-
Year 1
Year 2
Year 3

Figure 2

Conclusions: The present study indicates that the introduction of Cosyrel has two important effects:
1. It does not imply additional treatment costs; from the first year, actually, it generates a saving for the NHS.
2. The introduction of Cosyrel represents a benefit for the patients, especially for the elderly, because it improves the adherence to the therapy, thanks to the reduction in the number of pills taken. Better adherence is also linked to a reduction in the number of hospitalizations caused by therapy interruption and, consequently, permits to avoid the related costs.

1. World Health Organization in collaboration with the World Heart Federation and the World Stroke Organization. Global Atlas on cardiovascular disease prevention and control. Geneva, World Health Organization, 2011.
2. World Health Organization. *Global status report on non communicable diseases 2010.* Geneva, World Health Organization, 2011.
3. World Health Organization. The global burden of disease: 2004 update. Geneva, World Health Organization, 2004. Available at: www.who.int/healthinfo/global_burden_disease/2004_report_update/en/index.html.
4. Ambrosy AP, Fonarow GC, Butler J et al. The global health and economic burden of hospitalizations for heart failure. J Am Coll Cardiol 2014; 63(12): 1123-33.
5. Erdine S. How do compliance, convenience, and tolerability affect blood pressure goal rates? *Am J Cardiovasc Drugs* 2012; 12(5): 295-302.

MAProvider
MARKET ACCESS PROVIDER

via Carducci 24
20123 Milan - Italy
tel +39 02 89096682
fax +39 02 89098927
e-mail | info@maprovider.com

piazza San Salvatore in Lauro 10
00186 Rome - Italy
tel | fax +39 06 68806614
e-mail | info.roma@maprovider.com

web | www.maprovider.com

The quality management system operated by MA Provider S.r.l is in compliance with the standard UNI EN ISO 9001:2015 for planning and execution of consultancy services for the health care sector.